C2-565

AmpC β-Lactamase-Producing *Escherichia coli* from Canadian Hospitals: CANWARD 2007 & 2008

P.J. BAUDRY, S. PETERSON, **P. LAGACE-WIENS**, M.R. MULVEY, G.G. ZHANEL and D.J. HOBAN

University of Manitoba, Health Sciences Centre, Saint-Boniface General Hospital and the National Microbiology Laboratory, Winnipeg, Canada

Patricia Baudry
MS673-820 Sherbrook St.
Winnipeg, Manitoba, Canada
R3A 1R9
Phone: (204) - 787 - 4684
Fax: (204) - 787 - 4699
Email: trishbaudry@hotmail.com

REVISED ABSTRACT

Objective: The purpose of this study was to determine the prevalence and antimicrobial susceptibilities of AmpC β-lactamase-producing *E. coli* (AmpC-EC) from Canadian Hospitals in 2007 & 2008.

Methods: Clinically significant isolates were collected from January 2007 to December 2008 as part of the national CANWARD surveillance study. Susceptibility testing was performed by broth microdilution. Any *E. coli* with a cefoxitin (FOX) MIC ≥ 32 ug/ml was identified as a putative AmpC-hyperproducer and was screened for plasmidic *ampC* genes and for mutations within the chromosomal *ampC* promoter region by PCR and sequencing. Strains were typed using PFGE.

Results: 4.7% (76/1632) of *E. coli* was identified as AmpC-hyperproducers. Of the AmpC-EC, 55.3% were fluoroquinolone resistant, 36.8% were TMP/SMX resistant, 22.4% were gentamicin resistant and 36.8% were multidrug resistant (≥ 3 antimicrobial classes). All AmpC-EC remained susceptible to tigecycline and the carbapenems. 22.9% of isolates harbored bla_{CMY-2} and the remaining 76.1% displayed mutations in the chromosomal *ampC* promoter region previously shown to hyperproduce *ampC*. Genetically related (>80% homology) and unrelated groups of AmpC-EC were observed. Patient demographics revealed a mean age of 61 years with a range of 1 to 97 years, with females representing 56.6% of patients and males representing 43.4% of patients. AmpC-EC were isolated from across Canada with 23 (30.3%), 21 (27.6%), 19 (25.0%), 13 (17.1%) from BC/AB, QC/NS, ON and SK/MB, respectively. 36 (47.4%), 30 (39.5%), 7 (9.2%) and 3 (3.9%) of the AmpC-EC were isolated from blood, urine, wounds and respiratory, respectively.

Conclusion: The prevalence of AmpC-EC in Canadian hospitals is 4.7%. FOX resistance is mainly due to the hyperproduction of the chromosomal *ampC* and through acquisition of plasmidic bla_{CMY-2} gene. The spread of AmpC-EC in Canadian hospitals is extensive and is due to a combination of clonal spread and the dissemination of genetically unrelated strains.

INTRODUCTION

Escherichia coli is one of the most commonly encountered gram negative pathogens and causes a variety of community- and nosocomial-acquired infections. They generally remain susceptible to many classes of antimicrobials however there are increasing reports of resistance to the third generation cephalosporins (1, 2). Cephalosporin resistance is often mediated by the acquisition of extended-spectrum β -lactamases or by AmpC cephalosporinases.

AmpC β-lactamase genes belong to Ambler Class C and confer resistance to many β-lactam antimicrobials (excluding cefepime and carbapenems) (3).

In *E. coli* constitutive overexpression of AmpC β-lactamases can occur due to either the hyperproduction of the chromosomal AmpC β-lactamase gene or by acquisition of a plasmidic AmpC type β-lactamase (4). Plasmidic AmpC genes were first reported in 1988 and are now globally reported (3).

The purpose of this study was to determine the prevalence and antimicrobial susceptibilities of AmpC-EC identified from Canadian Hospitals in 2007 & 2008.

MATERIALS AND METHODS

Bacterial Isolates:

Bacterial isolates were collected as part of the **Can**adian **ward** (CANWARD) surveillance study. CANWARD is a laboratory based surveillance study coordinated by the Health Sciences Centre in Winnipeg, Canada. From January 2007 through December 2008, 12 sentinel Canadian hospitals (10 centres - 2008) submitted pathogens from patients attending hospital clinics, emergency rooms, medical and surgical wards, and intensive care units. Annually, each centre was asked to submit pathogens (consecutive, one per patient/infection site) from blood (360), respiratory (200), urine (100), and wound/IV (50) infections. 7881 and 5282 isolates were collected for 2007 and 2008, respectively. All isolates were identified at participating sites by routine procedures performed at each laboratory. Isolates were shipped to the reference laboratory at the Winnipeg Health Sciences Centre on Amies charcoal swabs, subcultured onto blood agar, and stocked in skim milk at -80°C until MIC testing was carried out.

Susceptibility Testing:

Following two subcultures from frozen stock, the *in vitro* activities of various antimicrobials were determined in duplicate by microbroth dilution in accordance with CLSI guidelines (M7-A7, 2006 and M100-S17, 2007). Colistin MICs were determined using the E-test method. Food and Drug Administration (FDA) interpretation breakpoints were used for tigecycline (S: ≤2 μg/mL, I: 4μg/mL and R: ≥8 μg/mL) and colisitn (S: ≤ 2 μg/mL and R: ≥4 μg/mL). Strains concomitantly resistant to ≥ 3 different antimicrobial classes were defined as multi-drug resistant (MDR). Any *E. coli* with a cefoxitin (FOX) MIC ≥ 32 ug/ml was identified as a putative AmpC-hyper producer.

Characterization of AmpC β-lactamase genes:

All isolates with an MIC of ≥ 32 ug/ml were screened for genes encoding EBC (ACT-1/MIR-1-related genes), DHA (DHA-related genes), FOX (FOX-related genes) and CIT (CMY-2-related genes) phylogenetic groups of AmpC acquired enzymes using a previously described multiplex PCR (6). A BLAST search of the DNA sequence was conducted to determine the specific gene. PCR for the amplification of the *ampC* promoter region was preformed as previously described (7).

Molecular Typing by PFGE

Genetic relationships of the AmpC hyperproducing *E. coli* were assessed by pulsed-field gel electrophoresis (PFGE) following digestion with *Xba*I as previously described (5).

REFERENCES

1. da Silva Dias, R. C. *et al.* 2008. Diagn. Microbiol. Infect. Dis. 60:79-87.
2. Goyanes, M. J. et al. 2007. Rev. Esp. Quimioter. 20: 216-221
3. Philippon, A. *et al*. 2004. Antimicrob. Agents Chemother. 46:1-11.
4. Woodford, N. et al. 2007. J. Antimicrob. Chemother. 59:102-105.
5. Mulvey, M. R. et al. 2005. Antimicrob. Agents Chemother.49: 358:365
6. Perez-Perez, F. J. et al. 2002. J. Clin. Microbiol. 40:2153-2162.
7. Caroff, N et al. 1999.FEMS Microbiol. Letters. 173:459-465.

RESULTS

Table 1: Prevalence of AmpC-producing *E. coli* in Canadian hospitals.

E. coli Isolates Submitted to CANWARD	% (n) of Isolates 2007	2008	2007/2008
AmpC-producers: Cefoxitin MIC ≥32 μg/ml	2.6% (13/500*)	5.6% (63/1132)	4.7% (76/1632)

* CANWARD 2007: Cefoxitin MICs were determined for 500 of 1702 *E. coli* collected

Table 2. Demographics of patients with infections due to AmpC-producing *E. coli* in Canadian hospitals.

Parameter	Value	Number (%) of Patients *E. coli* (n = 76)
Sex	Female	33 (43.4)
	Male	43 (56.6)
Age	≤ 17	6 (7.9)
	18 – 65	30 (39.5)
	≥ 66	40 (52.6)
Patient Region[a]	BC/AB (n = 2)*	23 (30.3)
	SK/MB (n = 2)*	13 (17.1)
	ONT (n = 4)*	19 (25.0)
	QC/Marit (n = 4, 2)*	21 (27.6)
Hospital Ward	Outpatient Clinics	10 (13.2)
	Emergency Rooms	28 (25.9)
	Intensive Care Units	9 (11.8)
	Medical Wards	26 (34.2)
	Surgical Wards	5 (6.6)
Specimen Type	Urine	30 (39.5)
	Blood	36 (47.4)
	Wound	7 (9.2)
	Respiratory Tract	3 (3.9)

*: Number of participating hospitals per region. QC/Marit: 4 centres in 2007 and 2 in 2008

Figure 1: Positions of mutations observed within the promoter and attenuater region of the chromosomal *ampC* gene

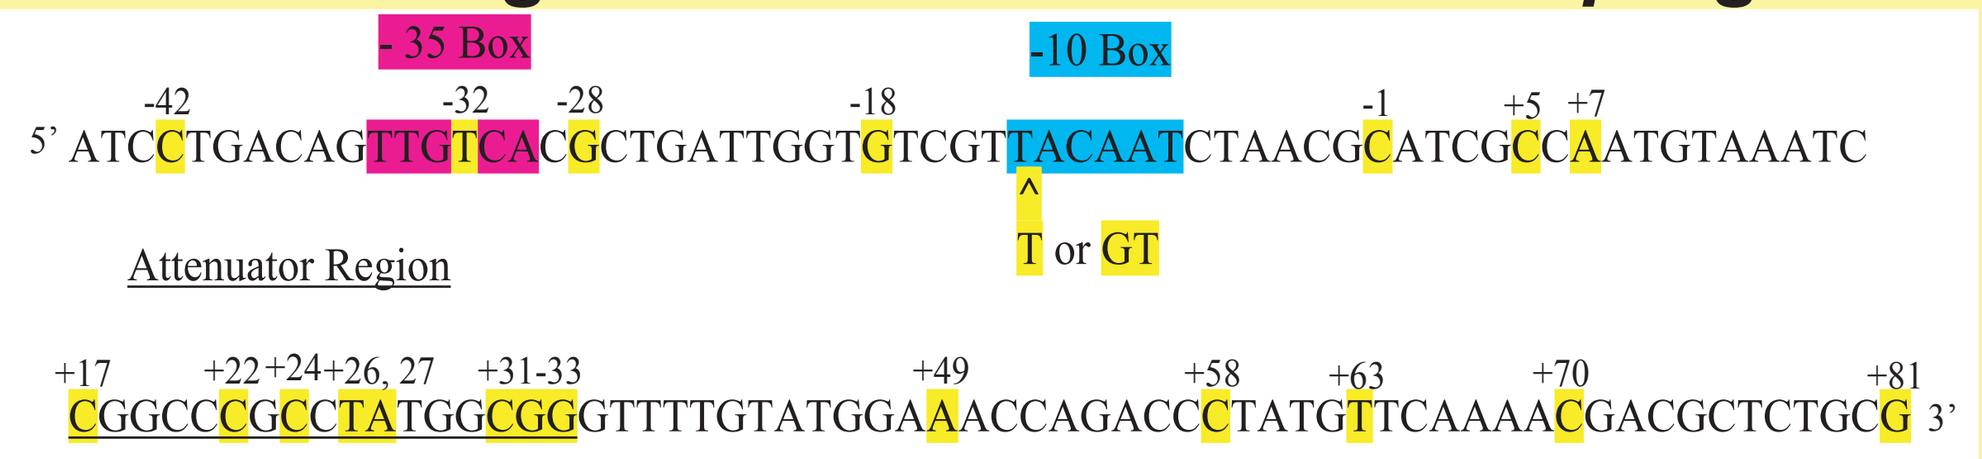
- 35 Box
-10 Box
-42 -32 -28 -18 -1 +5 +7
5' ATCCTGACAGTTGTCACGCTGATTGGTGTCGTTACAATCTAACGCATCGCCAATGTAAATC
^
T or GT
Attenuator Region
+17 +22 +24 +26, 27 +31-33 +49 +58 +63 +70 +81
CGGCCCGCCTATGGCGGGTTTTGTATGGAAACCAGACCCTATGTTCAAAACGACGCTCTGCG 3'

Figure 2: Genetic Relationships Among AmpC-producing *E. coli* from Canadian hospitals. PFGE using *Xba*I.

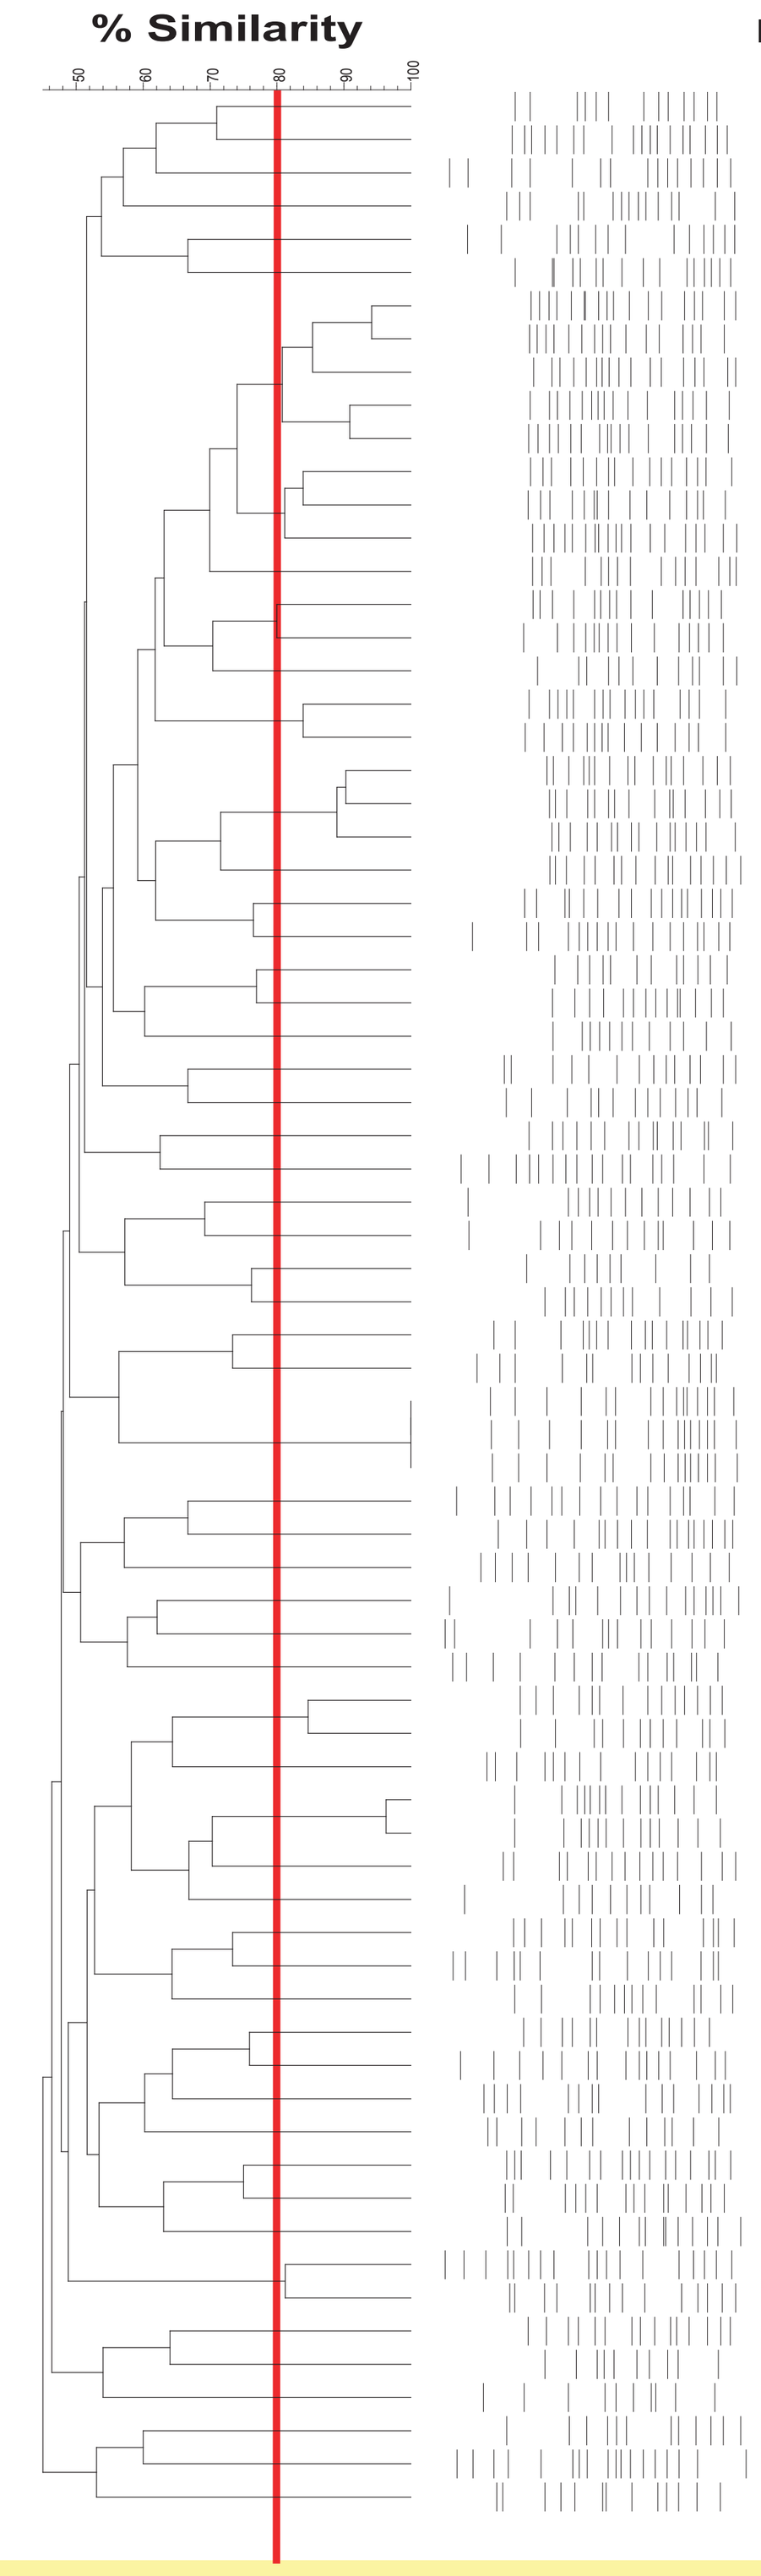
% Similarity
50
60
70
80
90
100

Isolate #	Region	Age	Sex	Genotype	Year
82376	SK/MB	90	M	-18 (G to A), -1 (C to T), +58 (C to T)	2008
82713	ONT	59	M	CMY-2	2008
78967	SK/MB	60	F	+22 (C to T), +26 (T to G), +27 (A to T), +32 (G to A), +70 (C to T)	2008
81196	ONT	1	F	-28 (G to A), +17 (C to T), +81 (G to A)	2008
76897	MARITIMES	77	M	CMY-2	2007
80189	QC	53	F	+70 (C to T)	2008
77343	QC	72	F	-28 (G to A), +81 (G to A)	2007
80240	QC	82	F	-28 (G to A), +81 (G to A)	2008
78399	BC/AB	73	F	CTX-M-3, TEM-1, OXA-1/-28 (G to A), +81 (G to A)	2007
82948	BC/AB	88	F	-32 (T to A), -28 (G to A), -18 (G to T), +31 (C to T), +81 (G to A)	2008
82988	BC/AB	78	M	-28 (G to A), +81 (G to A)	2008
80056	ONT	62	F	CTX-M-15, TEM-1, OXA-1/-28 (G to A), +81 (G to A)	2008
82412	SK/MB	78	M	CTX-M-15, OXA-1/-42 (C to T), -28 (G to A), +81 (G to A)	2008
76838	ONT	55	M	CTX-M-15, TEM-1, OXA-1/-28 (G to A), +81 (G to A)	2007
84251	QC	80	F	CMY-2	2008
80208	QC	75	F	-28 (G to A), +81 (G to A)	2008
81063	SK/MB	74	M	-32(TtoA), -28(GtoA), -11(CtoT), +15(CtoT), +22(CtoA), +49(AtoG), +81(GtoA)	2008
82805	QC	73	F	CMY-2	2008
80379	BC/AB	34	F	-42 (C to T), -28 (G to A), +81 (G to A)	2008
82294	ONT	14	M	-28 (G to A), +81 (G to A)	2008
75527	MARITIMES	30	F	-28 (G to A), +81 (G to A)	2007
76399	QC	65	F	Ins T at -14 (GTTA to GTTTA), +6 (C to T), +81(G to A)	2007
82932	BC/AB	51	F	-12 (A to T), +81 (G to A)	2008
77378	SK/MB	85	F	Ins T at -14 (GTTA to GTTTA), +6 (C to T), +81(G to A)	2007
79270	ONT	84	F	CMY-2	2008
80894	BC/AB	89	F	CMY-2	2008
80081	ONT	86	F	-32(T to A), +70 (C to T)	2008
82940	BC/AB	44	F	+70 (C to T)	2008
83194	SK/MB	78	F	-32 (to 'A), -28 (G to A), +31 (C to T), +81 (G to A)	2008
79444	MARITIMES	94	F	CMY-2	2008
81060	SK/MB	49	M	-18 (G to A), -1 (C to T), +58 (C to T)	2008
80952	BC/AB	4	M	-32 (T to A), -28 (G to A), -18 (G to T), +31 (C to T), +81 (G to A)	2008
83341	QC	47	M	-28 (G to A), +81 (G to A)	2008
76325	SK/MB	81	M	+58 (C to T), +63 (T to C), +81 (G to A)	2007
81067	SK/MB	73	M	-32 (T to A), +18 (G to A)	2008
80386	BC/AB	72	M	+22 (C to T), +26 (T to G), +27 (A to T), +32 (G to A), +70 (C to T)	2008
83402	BC/AB	33	F	CTX-M-27/-32 (T to A), +70 (C to T)	2008
81037	SK/MB	6	F	-32 (T to A), +70 (C to T)	2008
81986	BC/AB	21	F	+22 (C to T), +26 (T to G), +27 (A to T), +32 (G to A), +70 (C to T)	2008
83018	ONT	59	F	Ins GT @ -13/14 (GTCGTT to GTCGTGTT), +70 (C to T)	2008
83027	ONT	31	F	Ins GT @ -13/14 (GTCGTT to GTCGTGTT), +70 (C to T)	2008
83030	ONT	55	F	Ins GT @ -13/14 (GTCGTT to GTCGTGTT), +70 (C to T)	2008
79471	MARITIMES	80	F	-28 (G to A)	2008
83154	ONT	84	F	CMY-2	2008
78320	BC/AB	46	M	CMY-2	2007
79660	QC	75	M	-28 (G to A), +58 (C to T), +81 (G to A)	2008
80851	BC/AB	26	F	CMY-2	2008
81960	BC/AB	64	M	+22 (C to T), +26 (T to G), +27 (A to T), +32 (G to A), +70 (C to T)	2008
82001	BC/AB	41	F	InsT@ -14 (GTTAtoGTTTA),+22(CtoT),+26(TtoG),+27(AtoT),+32(GtoA),+70(CtoT)	2008
82929	BC/AB	77	M	CTX-M-15, TEM-1, OXA-1/+22 (CtoT), +26 (TtoG), +27 (AtoT), +32(GtoA),+70(CtoT)	2008
78299	BC/AB	70	F	-28 (G to A), +81 (G to A)	2007
79031	QC	56	M	-28 (G to A), +58 (C to T), +81 (G to A)	2008
79541	QC	56	M	-28 (G to A), +58 (C to T), +81 (G to A)	2008
84081	MARITIMES	32	M	CMY-2	2008
77734	SK/MB	36	M	-28 (G to A), +58 (C to T), +81 (G to A)	2008
80057	ONT	81	F	CMY-2	2008
81981	BC/AB	82	F	+22 (C to T), +26 (T to G), +27 (A to T), +32 (G to A), +70 (C to T)	2008
80059	ONT	81	F	CMY-2, TEM-1	2008
80871	BC/AB	77	M	-28 (G to A), +34 (G to A), +58 (C to T), +81 (G to A)	2008
82395	SK/MB	54	M	CTX-M-14, TEM-1/+22 (CtoT), +26 (TtoG), +27 (AtoT), +32 (G toA), +70 (CtoT)	2008
76751	ONT	79	M	+22 (C to T), +26 (T to G), +27 (A to T), +32 (G to A), +70 (C to T)	2007
85143	ONT	80	F	CTX-M-14, TEM-1/+22 (CtoT), +26 (TtoG), +27 (AtoT), +32 (GtoA), +70 (CtoT)	2008
78368	BC/AB	97	F	CMY-2	2007
81756	QC	64	F	+70 (C to T)	2008
77298	QC	58	M	CMY-2	2007
79652	QC	86	M	CMY-2	2008
79692	QC	75	M	-28 (G to A), +81 (G to A)	2008
80083	ONT	F	34	CTX-M-15, OXA-1/-28 (G to A), +34 (G to A), +58 (C to T), +81 (G to A)	2008
80835	BC/AB	3	F	CMY-2	2008
80861	BC/AB	63	M	Ins GT at -13/14 (GTCGTT to GTCGTGTT), +58 (C to T), +63 (T to C)	2008
80010	ONT	68	M	TEM-12/+70 (C to T)	2008
83819	AB	95	M	+22 (C to T), +26 (T to G), +27 (A to T), +32 (G to A), +70 (C to T)	2008
79341	ONT	93	M	CMY-2	2008

Figure 3: Antimicrobial resistance profiles among AmpC-producing *E. coli*.

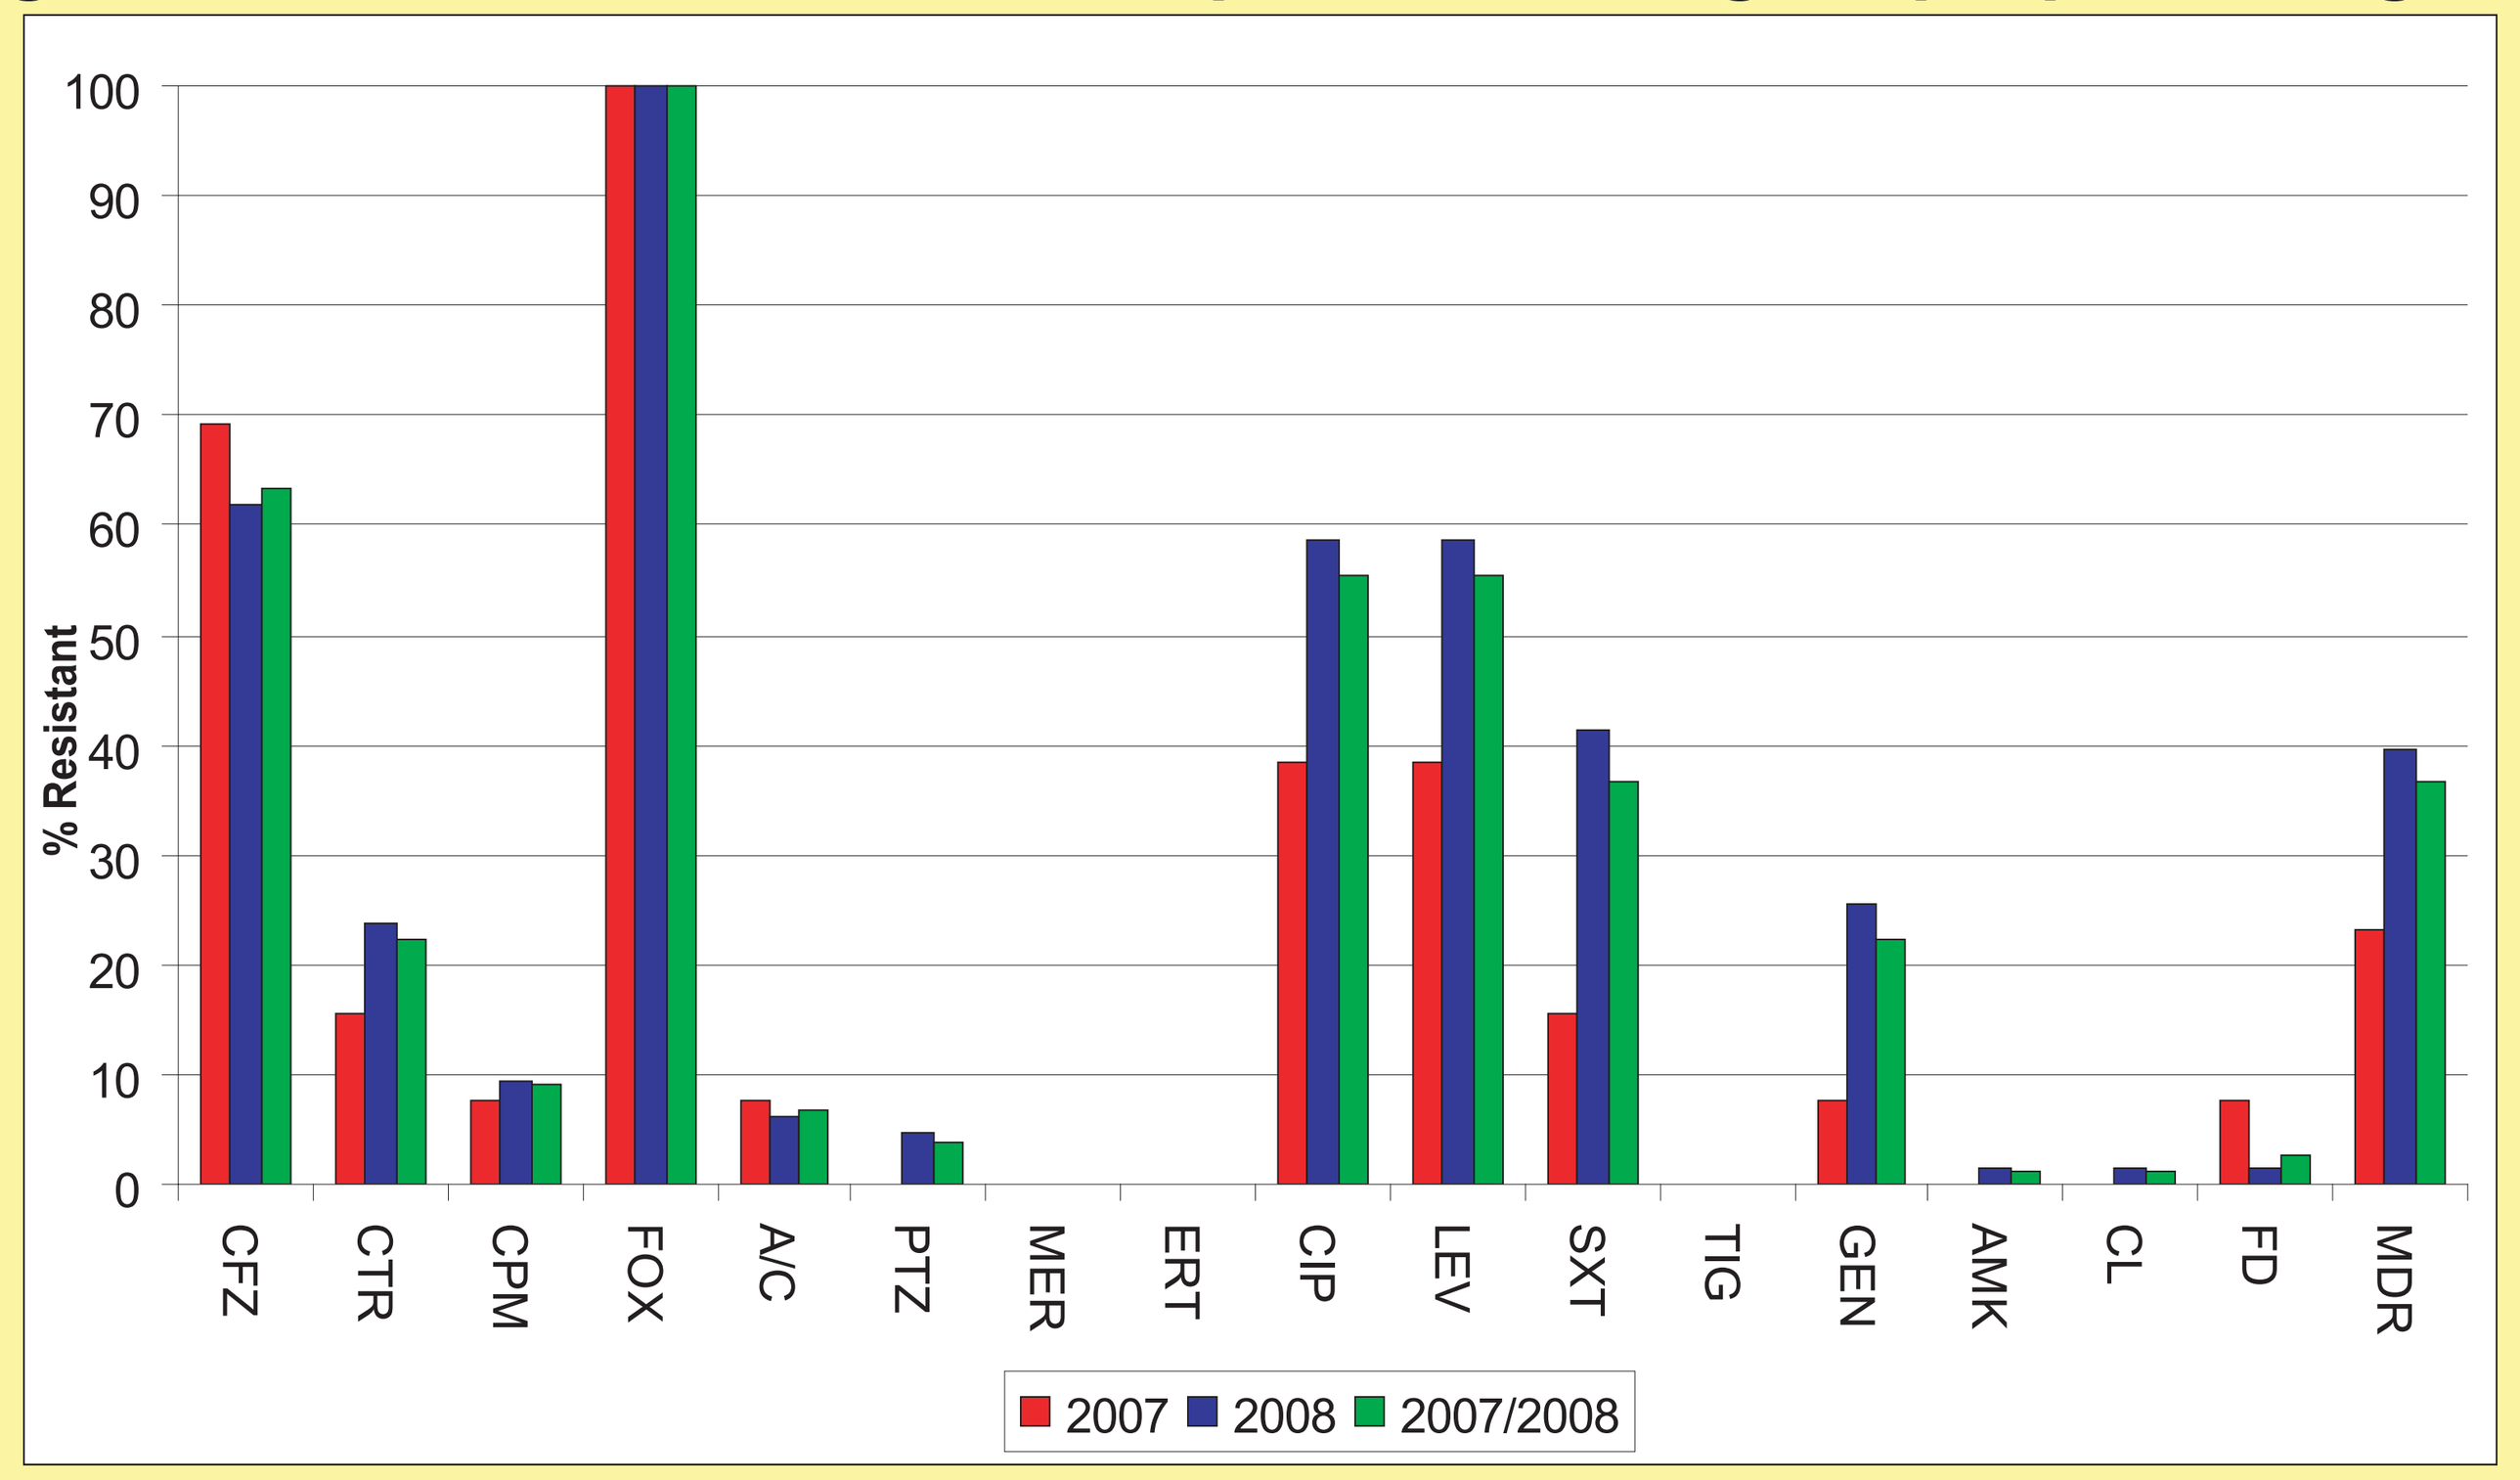
100
90
80
70
60
50
40
30
20
10
0
% Resistant
CFZ
CTR
CPM
FOX
A/C
PTZ
MER
ERT
CIP
LEV
SXT
TIG
GEN
AMK
CL
FD
MDR
2007
2008
2007/2008

ACKNOWLEDGEMENTS

Patricia Baudry is supported by the Natural Sciences and Engineering Research Council. The CANWARD study was supported in part by Abbott, Affinium Inc, Astellas, Bayer, Janssen Ortho Inc, Ortho McNeil, Merck, Pfizer, TaiGen, Targanta and Wyeth. The authors would like to thank the participating centres, investigators, and laboratory staff for their continued support.

CONCLUSIONS

1. The prevalence of AmpC-producing *E.coli* in hospital affiliated patients across Canada is 4.7%.
2. Multidrug-resistance was assocaited with 36.8% of AmpC-producing *E. coli.* However, they all remained susceptible to the carbapenems (meropenem and ertapenem) and tigecycline.
3. Cefoxitin resistance is mainly due to the hyperproduction of the chromosomal *ampC* gene (76.1%) and to a lesser extent through the acquisition of the plasmidic bla_{CMY-2} gene (22.9%).
4. The spread of AmpC-producing *E. coli* in Canadian hospitals is extensive and is due to a combination of clonal spread and the dissemination genetically unrelated strains.

Interscience Conference on Antimicrobial Agents and Chemotherapy- San Francisco, CA- Sept 12-15, 2009