C2-672

Extended-Spectrum β-Lactamase-Producing Enterobacteriaceae (ESBL-ENT) from Patients in Canadian Hospitals: CANWARD 2007 & 2008

P. J. BAUDRY, M. MCCRACKEN, **P. LAGACÉ-WIENS**, M.R. MULVEY, G.G. ZHANEL, D.J. HOBAN and the Canadian Antimicrobial Resistance Alliance (CARA).
University of Manitoba, Health Sciences Centre, Saint-Boniface General Hospital, and the National Microbiology Laboratory, Winnipeg, Canada

Patricia Baudry
MS673-820 Sherbrook St.
Winnipeg, Manitoba, Canada
R3A 1R9
Phone: (204) - 787 - 4684
Fax: (204) - 787 - 4699
Email: trishbaudry@hotmail.com

REVISED ABSTRACT

Objective: The purpose of this study was to assess the prevalence and resistance profiles of ESBL-ENT from patients in Canadian hospitals (CH) in 2007 & 2008.
Methods: Clinically significant isolates were collected from January 2007 to December 2008 as part of the CANWARD surveillance study. Susceptibility testing was performed by broth microdilution. Putative ESBL isolates were screened and confirmed following CLSI guidelines. PCR and DNA sequencing was used to detect resistance genes. Strains were typed using PFGE.
Results:

	% ESBL- *E. coli* (EC)	% ESBL- *K. pneumoniae* (KP)	% ESBL- *K. oxytoca* (KO)	% ESBL- *P. mirabilis* (PM)
2007	3.4 (53/1702)	1.6 (7/457)	0 (0/119)	0 (0/100)
2008	4.9 (55/1132)	3.2 (10/316)	0.8 (1/76)	0 (0/85)
2007 & 2008	3.8 (108/2834)	2.2 (17/773)	0.5 (1/195)	0 (0/185)

Resistance to fluoroquinolones, doxycycline, trimethoprim/sulfamethoxazole and gentamicin occurred in 82.5% and 58.8%, 75.9% and 70.6%, 71.3% and 70.6% and 54.6% and 52.9% of ESBL-EC and ESBL-KP, respectively. 83.3% of ESBL-EC, 76.5% of ESBL-KP and the 1 ESBL-KO were identified as multidrug resistant (MDR; ≥ 3 antimicrobial classes). The majority (>98- 100%) of ESBL-ENT remained susceptible to colistin, tigecycline and the carbapenems. CTX-M type was the predominant ESBL (80.3%), with CTX-M-15 as the predominant genotype (65.2%). 89.8% ESBL-ENT carried several β-lactamase genes. Generally the ENT harboring ESBLs were determined to be genetically unrelated (< 80% similarity) however small clusters of genetically related ESBL-EC were observed. Patient demographics were similar among ESBL-ENT.
Conclusions: The prevalence of ESBL-ENT in CH is increasing. *bla*$_{CTX-M-15}$ is the predominant genotype and has become endemic among ESBL-ENT which were frequently MDR. The spread of ESBL-ENT in CH is a combination of clonal spread and the dissemination of gentetically unrelated strains.

INTRODUCTION

Extended-spectrum β-lactamases (ESBLs) are the most significant resistance determinants emerging and spreading worldwide among the Enterobacteriaceae. They were first reported in 1983 and since then greater than 300 variants of ESBLs have been identified (http://www.lahey.org/Studies). The majority of ESBLs are Ambler Class A TEM, SHV and CTX-M variants. Until the late 1990's, TEM and SHV were the predominant ESBL variants and were mainly associated with nosocomial infections caused by *Klebsiella pneumoniae* (1). The epidemiology of ESBLs has undergone a rapid change over the last decade with the emergence and spread of CTX-M ESBLs. CTX-M ESBLs are now the most prevalent genotype and are mainly associated with *E. coli* infections from both community and nosocomial infections(2).

ESBLs are enzymes that compromise the efficacy of β-lactam antibiotics, except cephamycins and carbapenems, by hydrolysis of the β-lactam ring (1). Antimicrobial therapy is frequently limited for the treatment of ESBL-producers as they are often multi-drug resistant.

The purpose of this study was to assess the prevalence and resistance profiles of ESBL-producing Enterobacteriaceae from patients in Canadian hospitals in 2007 & 2008.

ACKNOWLEDGEMENTS

Patricia Baudry is supported by the Natural Sciences and Engineering Research Council. The CANWARD study was supported in part by Abbott, Affinium Inc, Astellas, Bayer, Janssen Ortho Inc, Ortho McNeil, Merck, Pfizer, TaiGen, Targanta and Wyeth. The authors would like to thank the participating centres, investigators, and laboratory staff for their continued support.

REFERENCES

1. Paterson, D. L. et al. 2005. Clin. Microbiol. Rev. 18: 657-686.
2. Canton, R. and Coque T. M. 2006. Curr Opin Microbiol 9:466-475.
3. Mulvey, M. R. et al. 2004. Antimicrob. Agents Chemother. 48:1204-1214.
4. Stewart, C.D. et al. 2001. J. Clin. Microbiol. 39:2864-2872.

MATERIALS AND METHODS

Bacterial Isolates:
Bacterial isolates were collected as part of the **Can**adian **ward** (CANWARD) surveillance study. CANWARD is a laboratory based surveillance study coordinated by the Health Sciences Centre in Winnipeg, Canada. From January 2007 through December 2008, 12 sentinel Canadian hospitals (10 centres - 2008) submitted pathogens from patients attending hospital clinics, emergency rooms, medical and surgical wards, and intensive care units. Annually, each centre was asked to submit pathogens (consecutive, one per patient/infection site) from blood (360), respiratory (200), urine (100), and wound/IV (50) infections. 7881 and 5282 isolates were collected for 2007 and 2008, respectively. All isolates were identified at participating sites by routine procedures performed at each laboratory. Isolates were shipped to the reference laboratory at the Winnipeg Health Sciences Centre on Amies charcoal swabs, subcultured onto blood agar, and stocked in skim milk at -80°C until MIC testing was carried out. In this study, only the analysis of unique ESBL-producing *Escherichia coli, Klebsiella pneumoniae, Klebsiella oxytoca* and *Proteus mirabilis* isolates will be reported.
Susceptibility Testing:
Following two subcultures from frozen stock, the *in vitro* activities of various antimicrobials were determined in duplicate by microbroth dilution in accordance with CLSI guidelines (M7-A7, 2006 and M100-S17, 2007). Colistin MICs were determined using the E-test method. Food and Drug Administration (FDA) interpretation breakpoints were used for tigecycline (S: ≤2 µg/mL, I: 4µg/mL and R: ≥8 µg/mL) and colisitn (S:≤ 2µg/mL and R: ≥4µg/mL). Strains concomitantly resistant to ≥ 3 different antimicrobial classes were defined as multi-drug resistant (MDR). Any *E. coli, K. penumoniae, K. oxytoca and P. mirabilis* with a ceftriaxone and/or a ceftazidime MIC ≥ 1 µg/mL was identified as a putative ESBL and underwent further analysis. The putative ESBL phenotype was confirmed by the disk confirmation method as described by CLSI. *E. coli* ATCC 25922 and *Klebsiella pneumoniae* ATCC 700603 were the control strains used in this study.
Characterization of ESBLs
Genotypic characterization of ESBLs was performed by PCR and sequencing of *bla*$_{SHV}$, *bla*$_{TEM}$, *bla*$_{CTX-M}$, and *bla*$_{OXA}$ genes as previously described (3, 4). A BLAST search of the DNA sequence was conducted to determine specific genotypes.
Molecular Typing by PFGE
Genetic relationships of the ESBL-producing *E. coli* and *K. pneumoniae* were assessed by pulsed-field gel electrophoresis (PFGE) following digestion with *Xba*I as previously described (3).

CONCLUSIONS

1. The prevalence of ESBL-producing Enterobacteriaceae in hospital affiliated patients across Canada is increasing.
2. The prevalence of ESBL-producing *E. coli* and *K. pneumoniae* is highest among surgical and medical wards in Canadian hospitals and is most commonly isolated from blood and urine specimen.
3. Multi-drug resistance was found to be associated with 83.3% of ESBL-producing *E. coli* and 76.5% of ESBL-producing *Klebsiella pneumoniae*.
4. The majority (>98- 100%) of ESBL-ENT remained susceptible to the carbapenems (ertapenem & meropenem), colistin (polymyxin E), and tigecycline.
5. CTX-M was the predominant ESBL type (80.3%) with *bla*$_{CTX-M-15}$ (65.2%) as the predominant genotype among ESBL-producing Enterobacteriaceae.
6. Based on PFGE, the spread of ESBL-producing Enterobacteriaceae across Canada is a combination of clonal spread and the dissemination genetically unrelated strains.

RESULTS

Figure 1: Prevalence of ESBL-producing Enterobacteriaceae in Canadian hospitals.

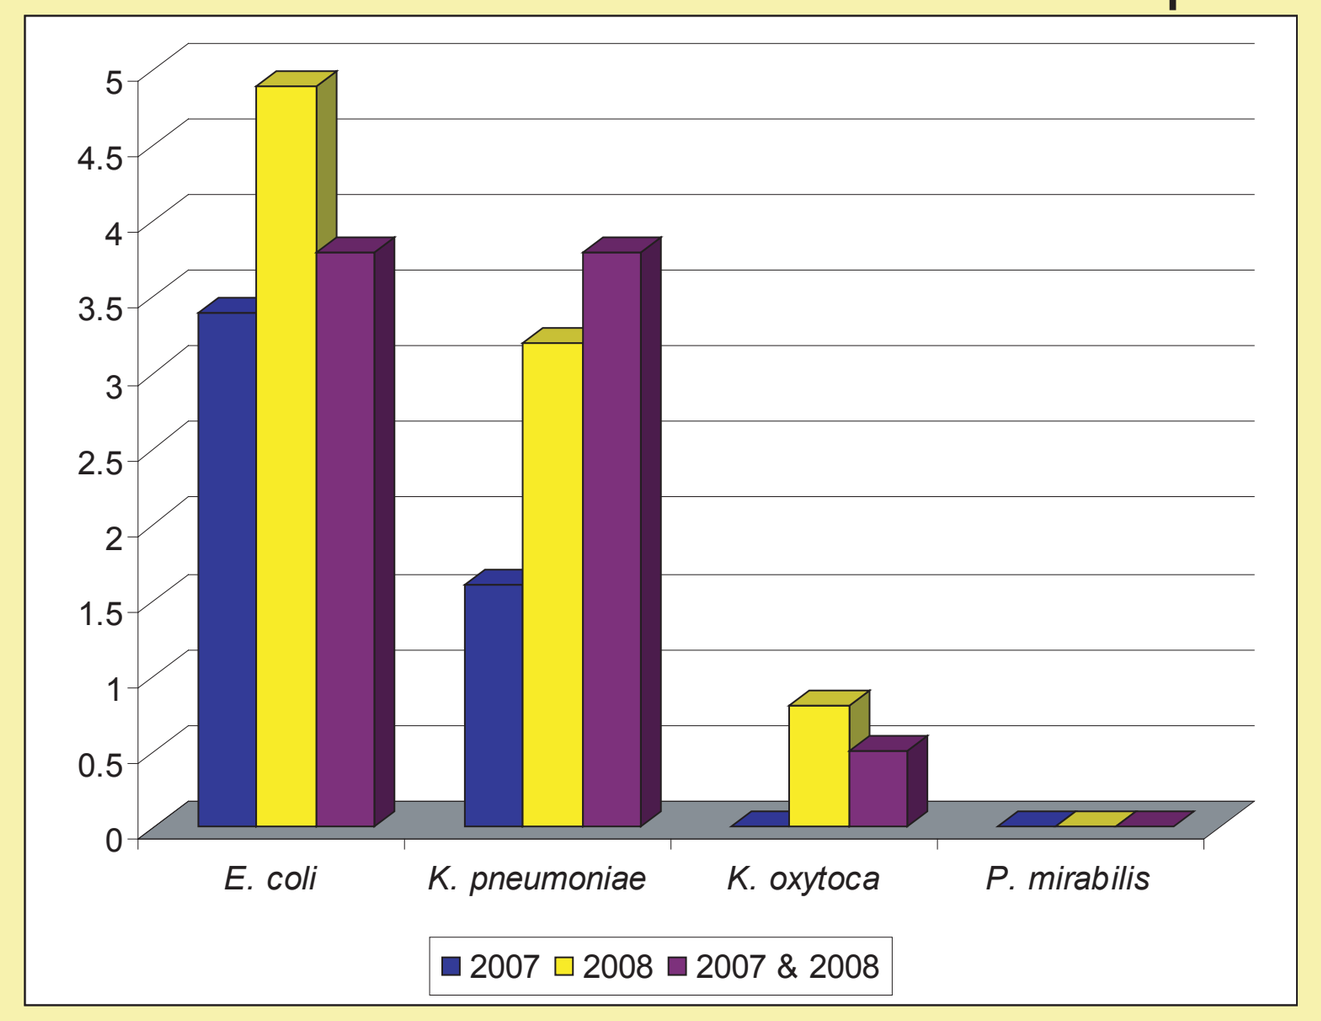
5
4.5
4
3.5
3
2.5
2
1.5
1
0.5
0
E. coli
K. pneumoniae
K. oxytoca
P. mirabilis
2007
2008
2007 & 2008

Figure 2: Prevalence of ESBL-producing Enterobacteriaceae by hospital ward.

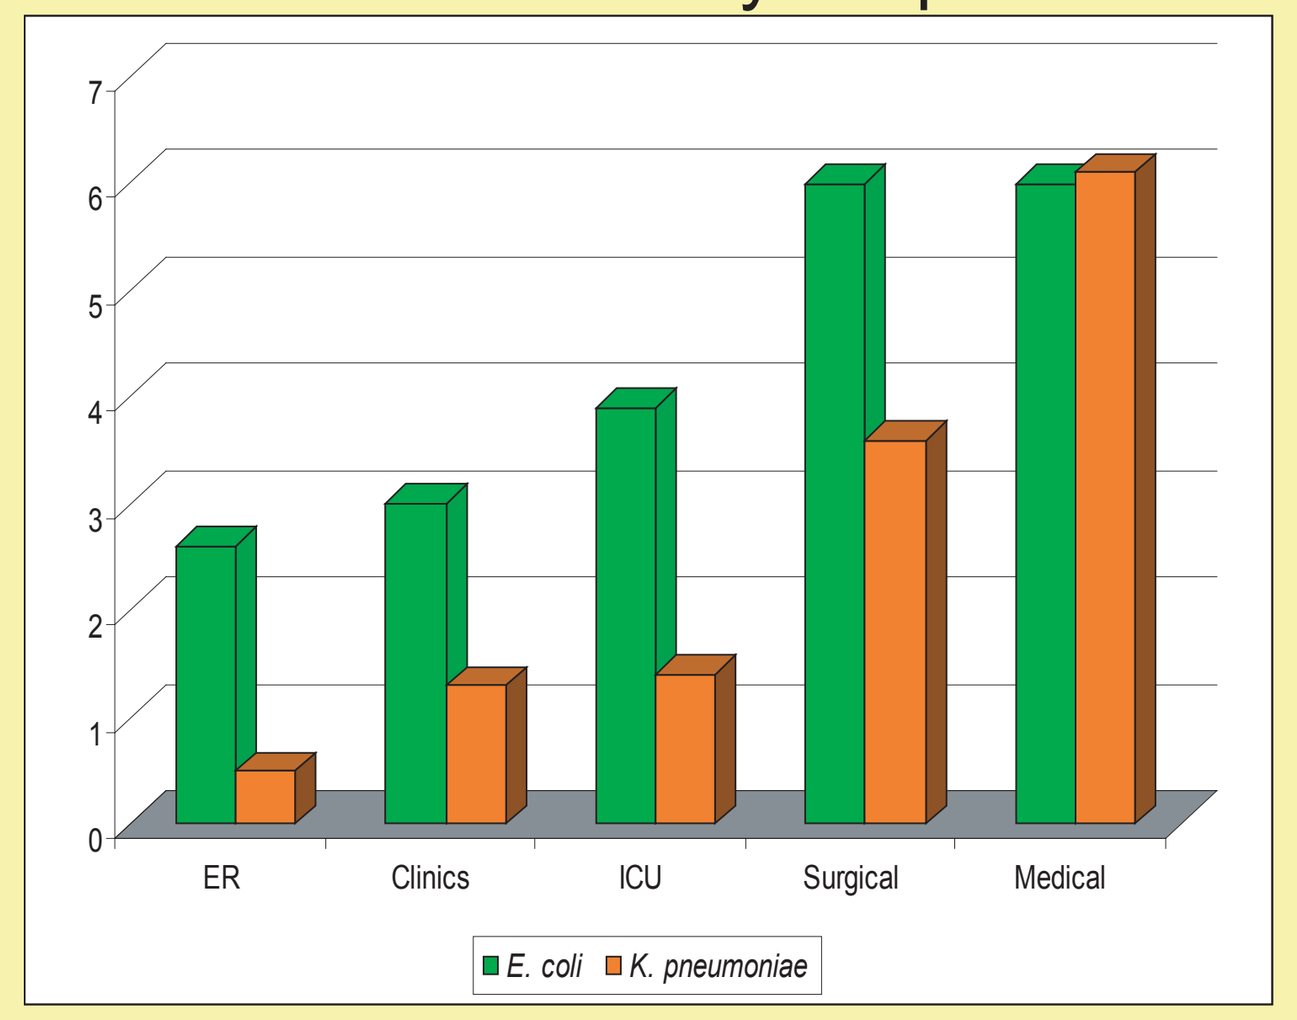
7
6
5
4
3
2
1
0
ER
Clinics
ICU
Surgical
Medical
E. coli
K. pneumoniae

Figure 3: PFGE dendrogram depicting the genetic relationships among ESBL-producing *E. coli*.

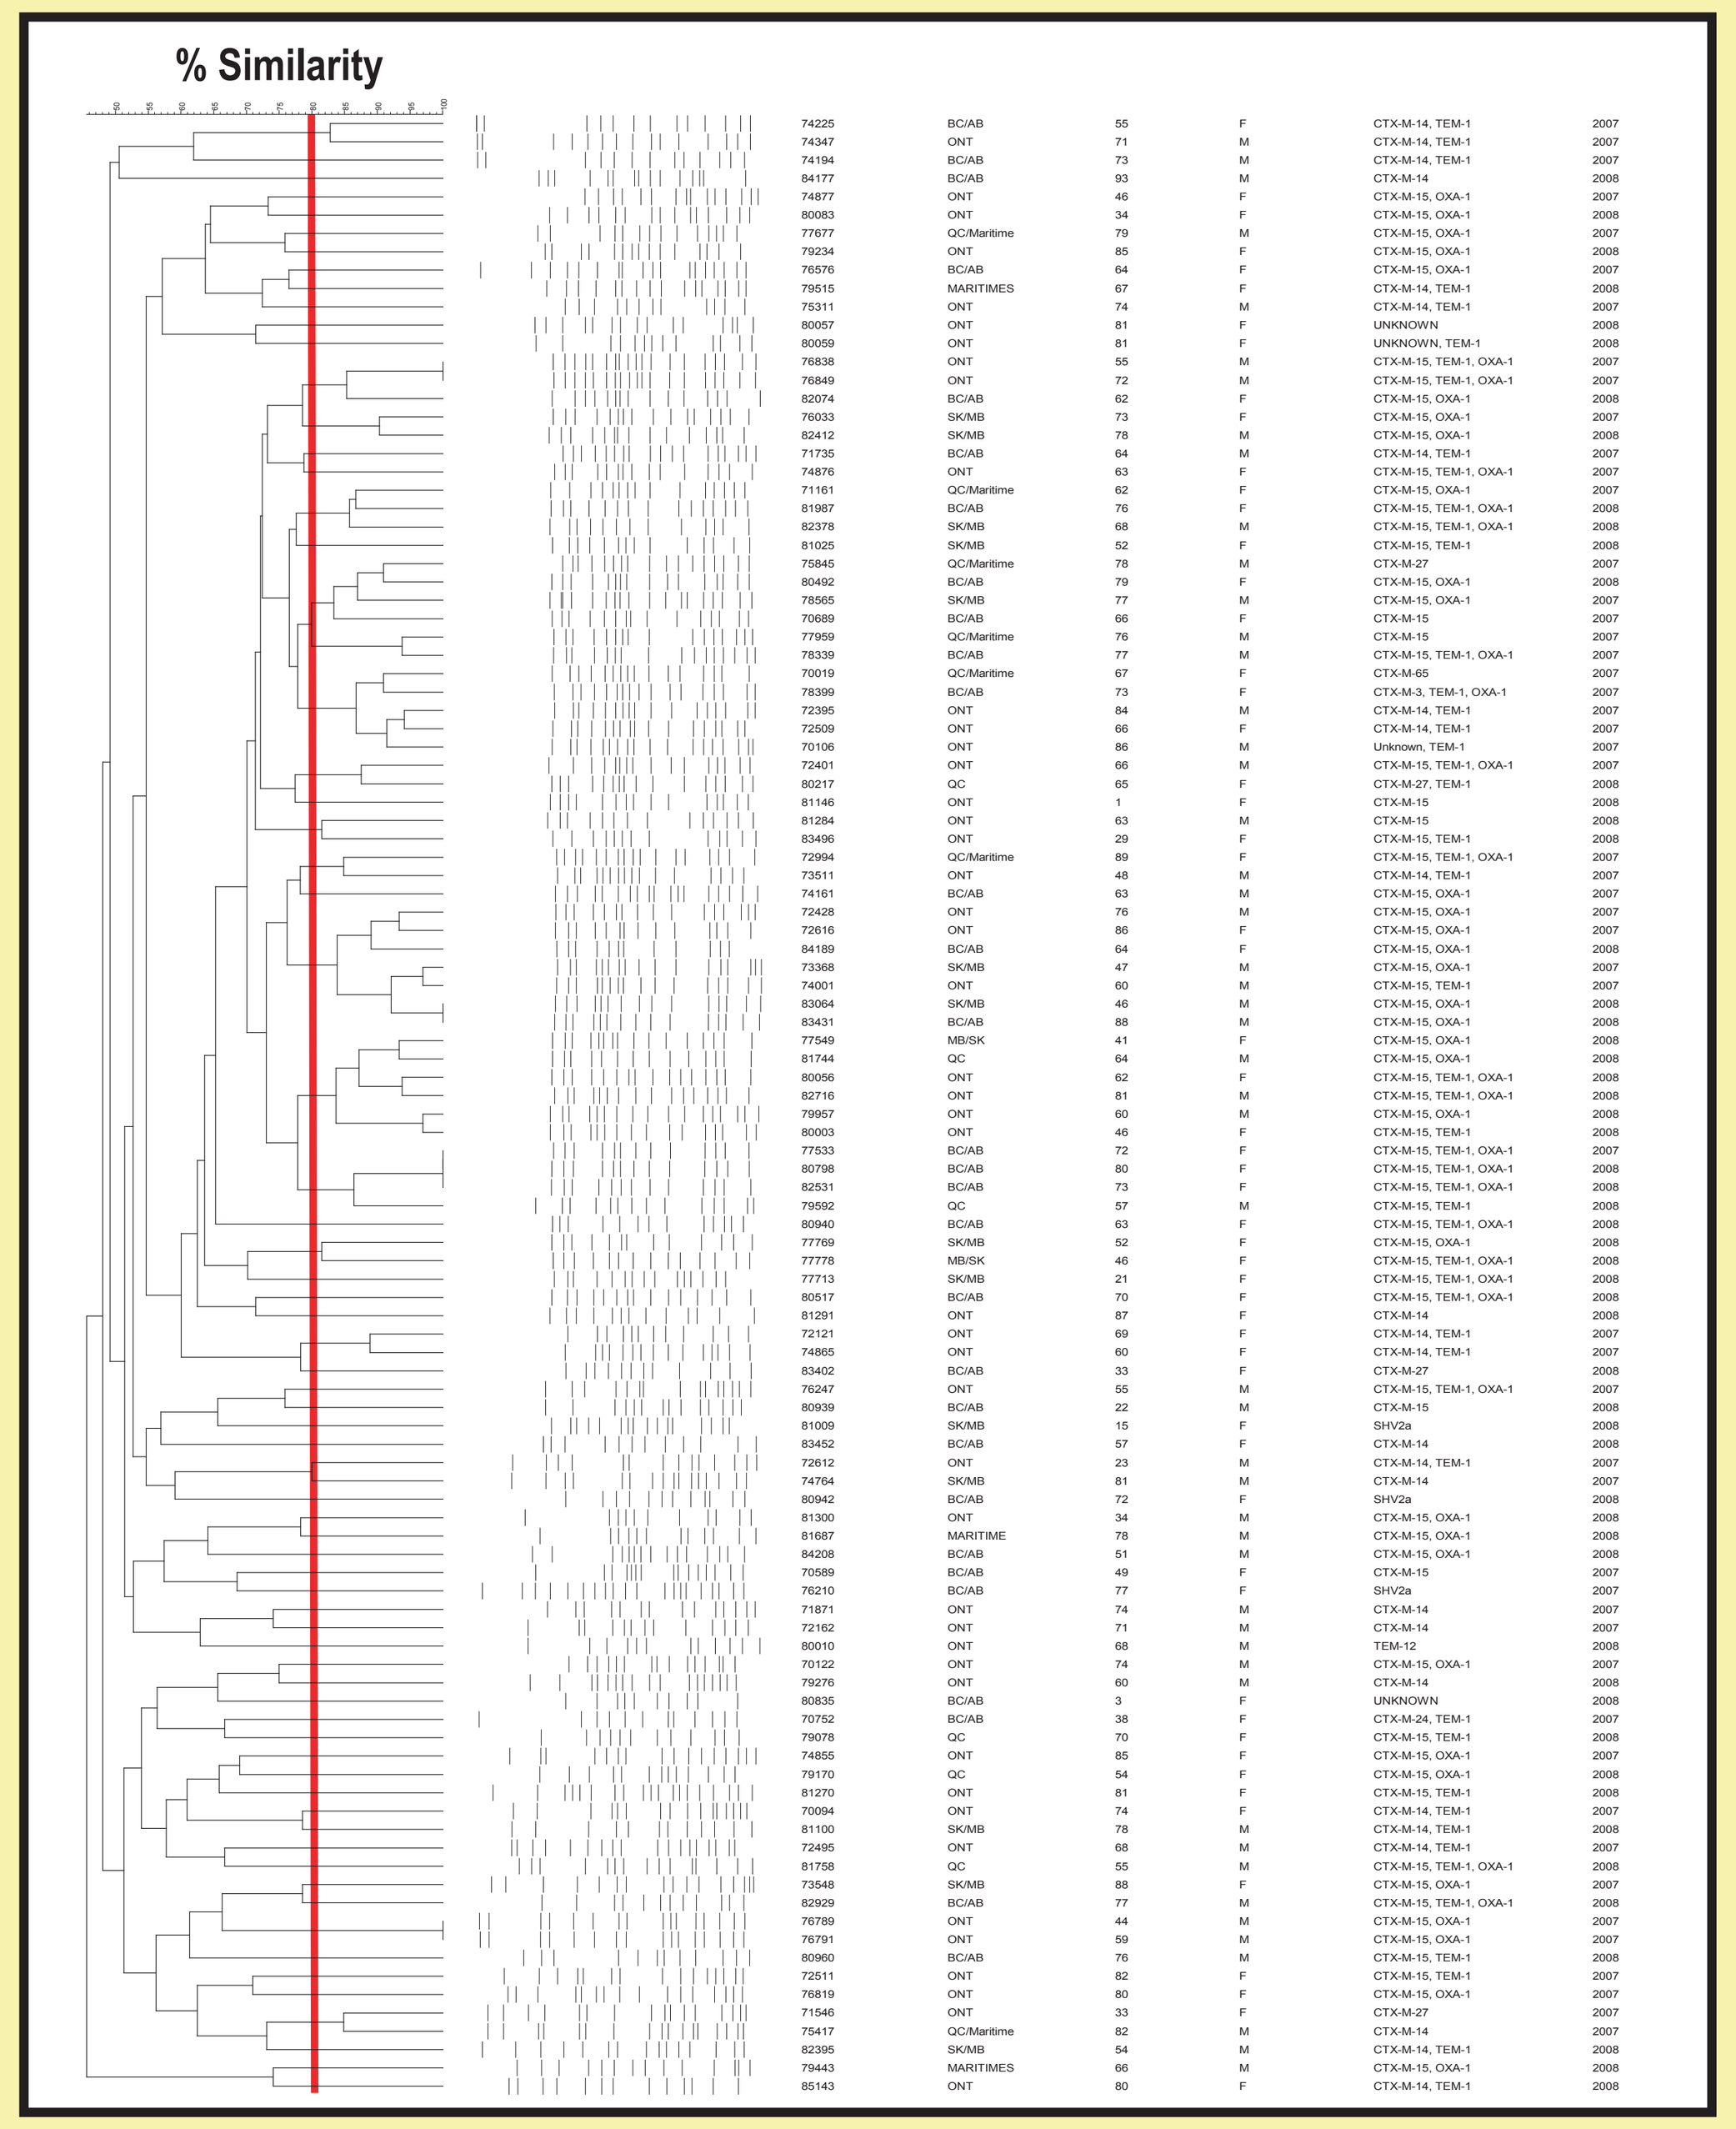
% Similarity

Table 1: Demographics of patients with infections due to ESBL-producing *E. coli and K. pneumoniae* in Canadian hospitals.

Parameter	Value	Number (%) of Patients: *E. coli* (n = 108)	Number (%) of Patients: *K. pneumoniae* (n = 17)
Sex	Female	57 (52.8)	7 (41.2)
	Male	51 (47.2)	10 (58.8)
Age	≤ 17	3 (2.8)	1 (5.9)
	18 – 65	45 (41.7)	9 (52.9)
	≥ 66	60 (55.6)	7 (41.2)
Patient Region [a]	BC/AB (n = 2)*	30 (27.8)	4 (23.5)
	SK/MB (n = 2)*	16 (14.8)	1 (5.9)
	ONT (n = 4)*	46 (42.6)	8 (47.1)
	QC/Marit (n = 4, 2)*	16 (14.8)	4 (23.5)
Patient Type	In	65 (60.2)	15 (88.2)
	Out	43 (39.8)	2 (11.8)
Hospital Ward	Outpatient Clinics	12 (11.1)	1 (5.9)
	Emergency Rooms	28 (25.9)	1 (5.9)
	Intensive Care Units	9 (8.3)	2 (11.8)
	Medical Wards	48 (44.4)	10 (58.8)
	Surgical Wards	11 (10.2)	3 (17.6)
Specimen Type	Urine	39 (36.1)	6 (35.3)
	Blood	62 (57.4)	7 (41.2)
	Wound	2 (1.9)	2 (11.8)
	Respiratory Tract	5 (4.6)	2 (11.8)

*: Number of participating hospitals per region. QC/Marit: 4 centres in 2007 and 2 in 2008

Table 2: Distribution of β-lactamase genes among ESBL-producing *E. coli* and *K. pneumoniae*.

Genotype*	Organism	Number of isolates
CTX-M-2, SHV-11	*K. pneumoniae*	1
CTX-M-3, TEM-1, OXA-1	*E. coli*	1
CTX-M-14	*E. coli*	7
CTX-M-14, TEM-1	*E. coli*	18
CTX-M-14, TEM-1, SHV-11	*K. pneumoniae*	1
CTX-M-15	*E. coli*	6
CTX-M-15, TEM-1	*E. coli*	10
CTX-M-15, OXA-1	*E. coli*	33
CTX-M-15, SHV-1, OXA-1	*K. pneumoniae*	1
CTX-M-15, SHV-28, OXA-1	*K. pneumoniae*	1
CTX-M-15, TEM-1, OXA-1	*E. coli*	19
CTX-M-15, TEM-1, SHV-5, OXA-1	*K. pneumoniae*	1
CTX-M-15, TEM-1, SHV-11, OXA-1	*K. pneumoniae*	2
CTX-M-15, TEM-1, SHV-1	*K. pneumoniae*	1
CTX-M-15, TEM-1, SHV-11	*K. pneumoniae*	1
CTX-M-24, TEM-1	*E. coli*	1
CTX-M-27	*E. coli*	3
CTX-M-27, TEM-1	*E. coli*	1
CTX-M-65	*E. coli*	1
SHV2a	*E. coli*	3
SHV2a	*K. pneumoniae*	2
SHV2a, OXA-1	*K. pneumoniae*	1
SHV-12, TEM-1	*K. pneumoniae*	1
TEM-12	*E. coli*	1
Unknown	*E. coli*	2
Unknown, SHV-1	*K. pneumoniae*	4
Unknown, TEM-1	*E. coli*	2

Figure 4: PFGE dendrogram depicting the genetic relationships among ESBL-producing *K. pneumoniae*.

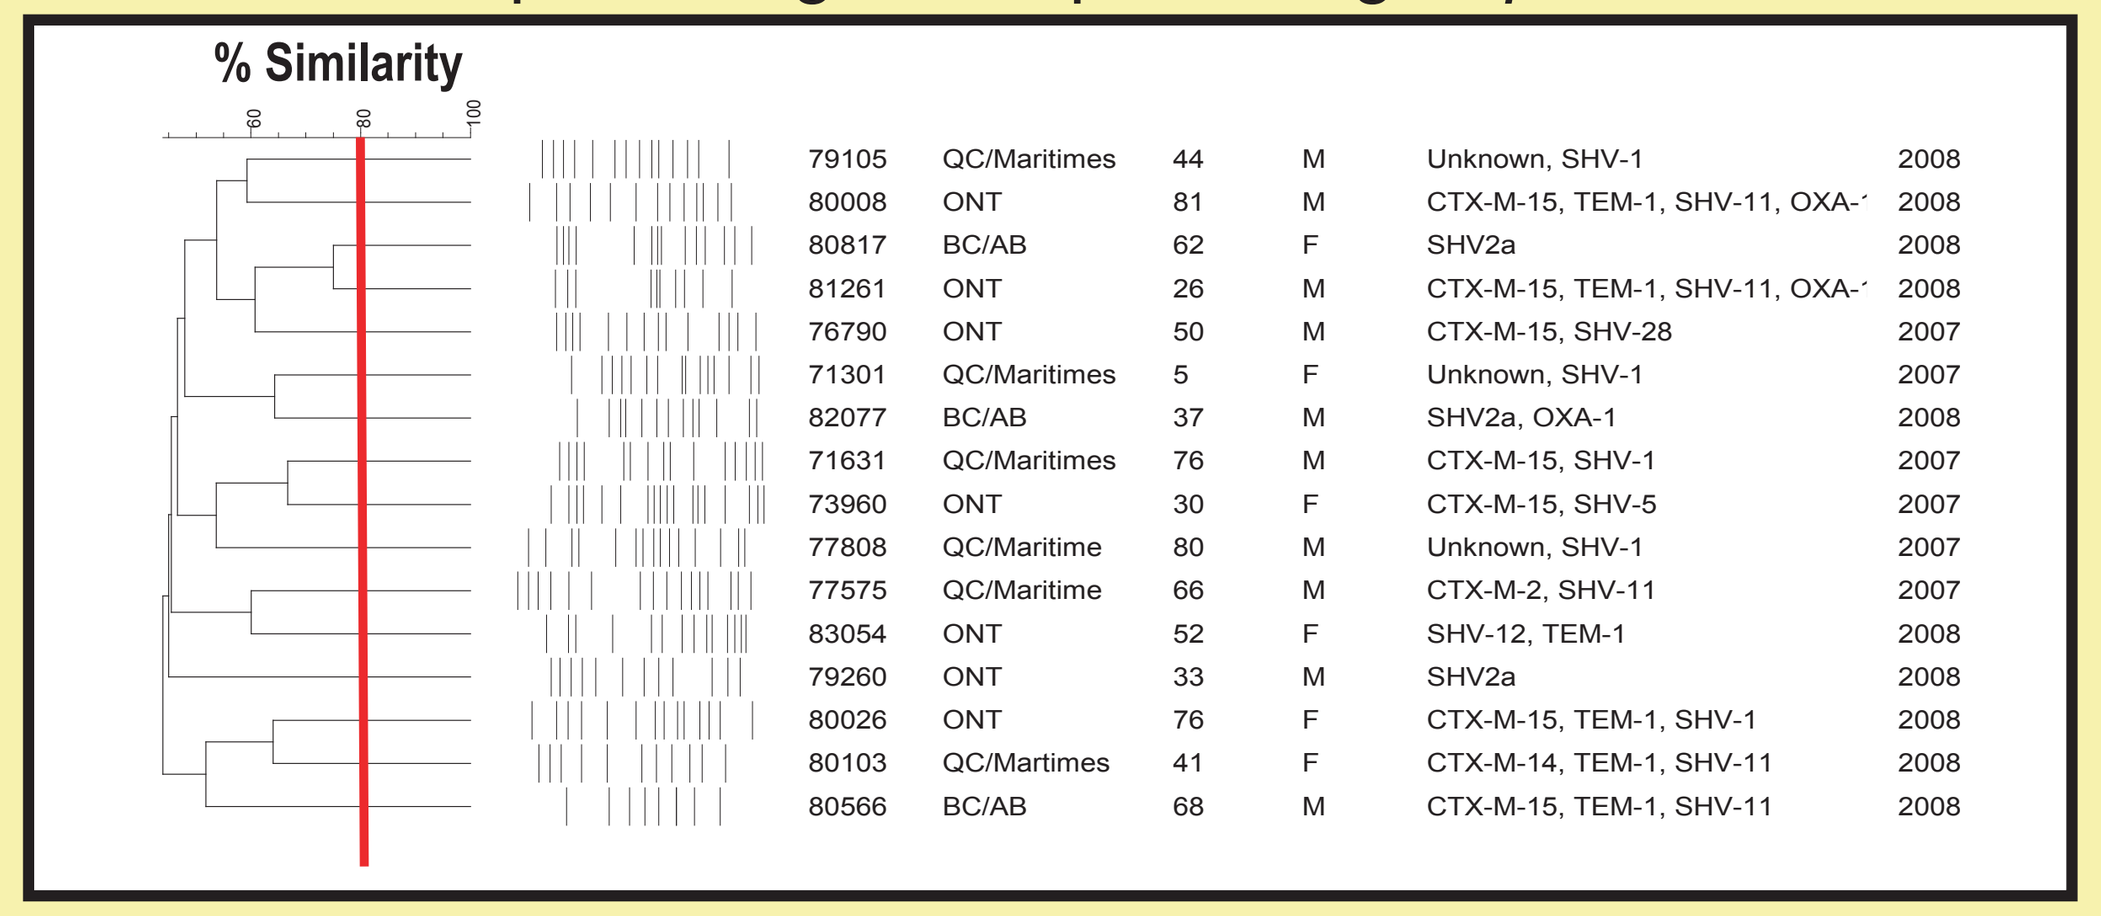
% Similarity
79105 QC/Maritimes 44 M Unknown, SHV-1 2008
80008 ONT 81 M CTX-M-15, TEM-1, SHV-11, OXA-1 2008
80817 BC/AB 62 F SHV2a 2008
81261 ONT 26 M CTX-M-15, TEM-1, SHV-11, OXA-1 2008
76790 ONT 50 M CTX-M-15, SHV-28 2007
71301 QC/Maritimes 5 F Unknown, SHV-1 2007
82077 BC/AB 37 M SHV2a, OXA-1 2008
71631 QC/Maritimes 76 M CTX-M-15, SHV-1 2007
73960 ONT 30 F CTX-M-15, SHV-5 2007
77808 QC/Maritime 80 M Unknown, SHV-1 2007
77575 QC/Maritime 66 M CTX-M-2, SHV-11 2007
83054 ONT 52 F SHV-12, TEM-1 2008
79260 ONT 33 M SHV2a 2008
80026 ONT 76 F CTX-M-15, TEM-1, SHV-1 2008
80103 QC/Martimes 41 F CTX-M-14, TEM-1, SHV-11 2008
80566 BC/AB 68 M CTX-M-15, TEM-1, SHV-11 2008

Figure 5: Antimicrobial resistance profiles among ESBL-producing *E. coli*.

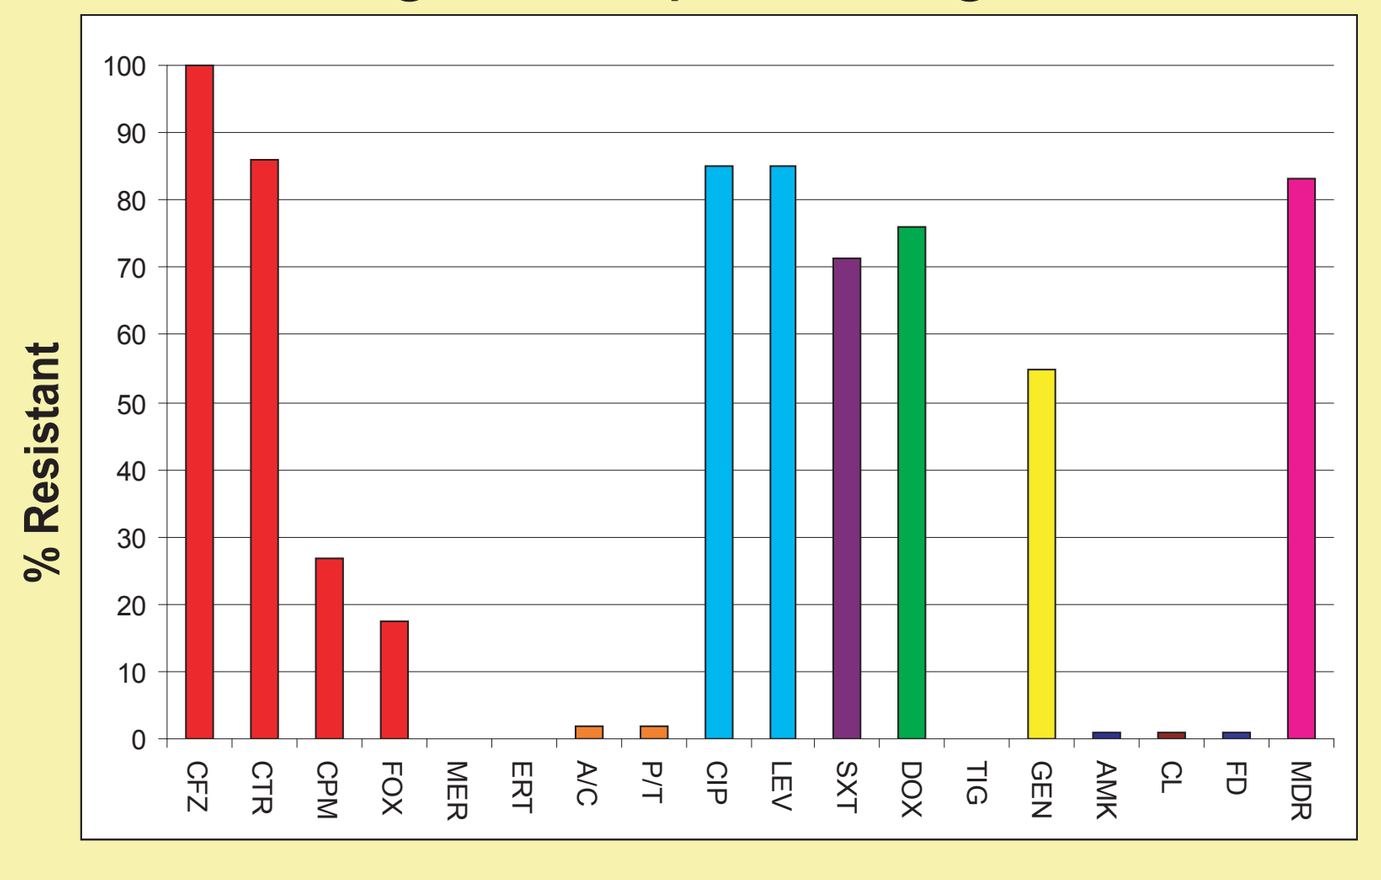
% Resistant
100
90
80
70
60
50
40
30
20
10
0
CFZ
CTR
CPM
FOX
MER
ERT
A/C
P/T
CIP
LEV
SXT
DOX
TIG
GEN
AMK
CL
FD
MDR

Figure 6: Antimicrobial resistance profiles among ESBL-producing *K. pneumoniae*.

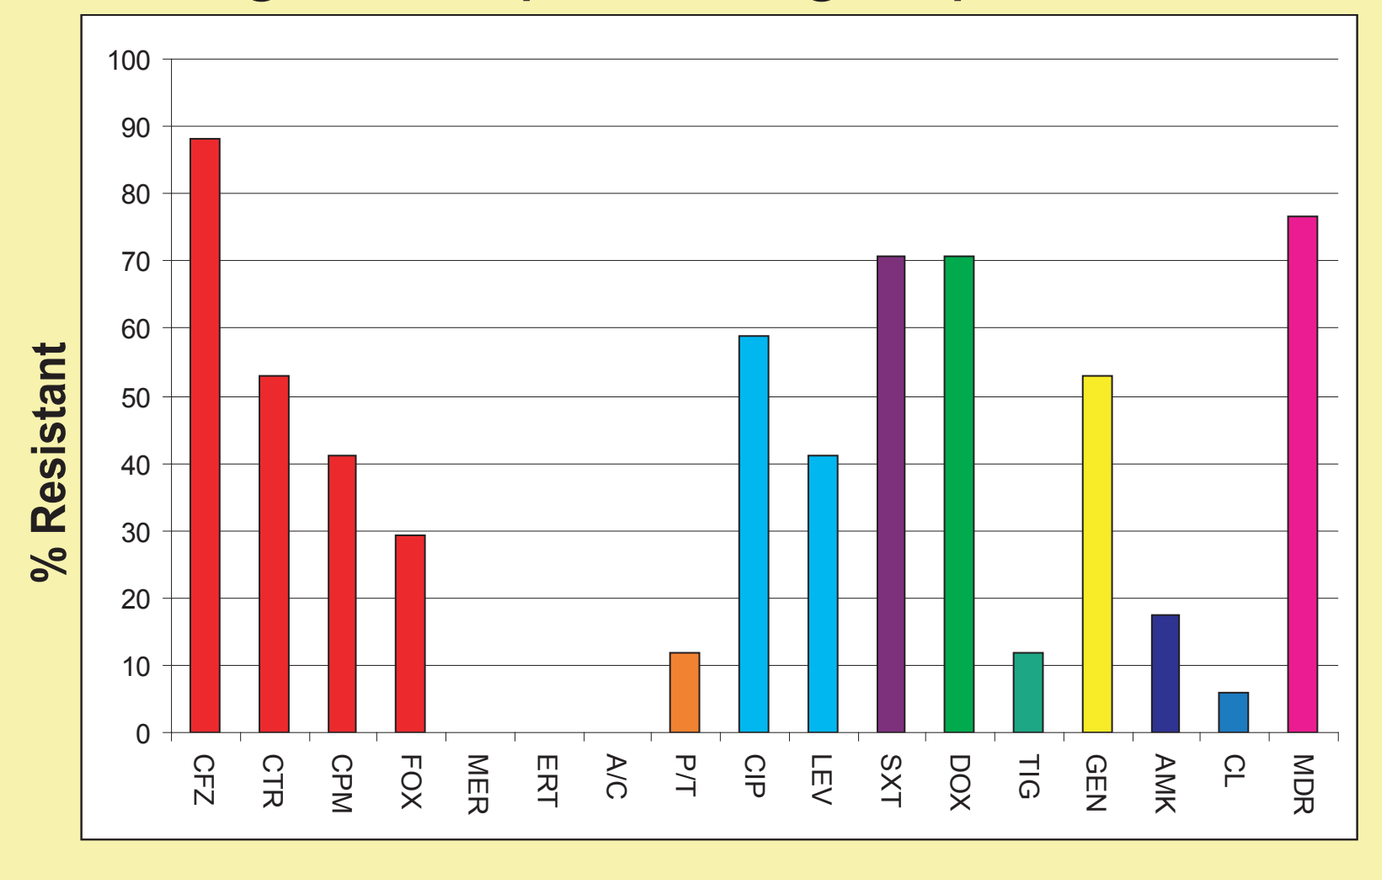
% Resistant
100
90
80
70
60
50
40
30
20
10
0
CFZ
CTR
CPM
FOX
MER
ERT
A/C
P/T
CIP
LEV
SXT
DOX
TIG
GEN
AMK
CL
MDR

CFZ: cefazolin, CTR: ceftriaxone, CPM: cefepime, FOX: cefoxitin, MER: meropenem, ERT: ertapenem, A/C: amoxicillin/clavulanic acid, PTZ: piperacillin/tazobactam, CIP: ciprofloxacin, LEV: levofloxacin, SXT: trimethoprim/sulfamethoxazole, DOX: doxycycline, TIG: tigecycline, GEN: gentamicin, AMK: amikacin, CL: colistin, FD: nitrofurantoin, MDR: multi-drug resistant

Interscience Conference on Antimicrobial Agents and Chemotherapy - San Francisco, CA - September 12-15, 2009